Evaluation of Different Sample Preparations and Application of a Novel Surfactant for Peptide Analysis in Biological Matrices Using Online SPE-LC/MS/MS

Yan Wang[1], Isabelle Tcholakov[1], Michel Koch[2], Miryam Kadkhodayan[1]
[1]Amylin Pharmaceuticals, Inc., San Diego, CA; [2]Spark Holland BV, Emmen, The Netherlands

Overview

Purpose

- Evaluation 2 new extraction techniques: an anion exchange and a reverse-phase extraction procedure for peptide analysis in biological matrices using online SPE-LC/MS
- Examination PPS Silent Surfactant for quenching enzymatic activity and enhancing sample clean-up in sample preparation for peptide analysis

Instrumentation

- Applied Biosystems API 3000™ LC-MS/MS system
- Symbiosis Pharma™ (integrated HPLC pumps, autosampler and online SPE)

Results

- Anion exchange procedure using Oasis® MAX provided cleaner sample preparation in biological matrices
- PPS at concentration of 0.1% effectively stopped the enzymatic activity of human plasma
- Application of 0.1% PPS improved extraction recovery; however, it caused increased % CV in the data of triplicate samples

Introduction

- Sample preparation for analytes in biological matrices is a critical part of successful LC MS/MS analyses
 - Analysis of larger peptides is particularly challenging due to some unique properties, such as non-specific surface interaction and poor solubility, which can affect extraction yields
 - These properties can severely impact the recovery of large peptides from biological matrices
- This work evaluated 2 new extraction techniques: an anion exchange procedure using Oasis MAX (Waters Corporation, USA) and a reverse-phase extraction procedure using HySphere GP (Spark Holland, The Netherlands)
- Comparisons were made utilizing an online SPE–LC/MS/MS. In addition, PPS Silent™ Surfactant (Protein Discovery, USA) was examined as a mass spectrometry-compatible detergent for quenching enzymatic activity and enhancing sample clean-up

Materials and Methods

- Samples were prepared by adding a peptide to pooled human plasma
 - Prior to extraction, all plasma samples were diluted 1:4 with either PPS (final concentration 0.1 to 1%) or aqueous solutions (acid/base)
 - Diluents were 1% formic acid, 5% H_3PO_4, and 5% NH_4OH for GP cartridges and MAX, respectively.
- Diluted sample (50 µL) was injected onto a system capable of performing online solid phase extraction
 - Samples were subsequently eluted from the cartridge onto a reverse-phase analytical column (Guard Column, Michrom Bioresources/RP Polymeric 1 x 20 mm) coupled to an SCIEX API 3000 mass spectrometer equipped with an ESI source

Table 1. HPLC gradient method

Time (min:sec)	Flow Rate (mL/min)	% Solvent A	% Solvent B
0:00	0.6	95	5
1:00	0.6	95	5
1:10	0.7	95	5
2:10	0.7	5	95
2:40	0.7	5	95
2:50	0.7	95	5
3:50	0.7	95	5

Solvent A: Water/0.05% TFA/0.005% HFBA; Solvent B: ACN/0.05% TFA/0.005% HFBA.

Table 2. Solid-phase Oasis MAX extraction conditions

#	Step	Solvent	Flow Rate (µL/min)	Volume (µL)
1	SPE Cartridge Activation	MeOH	5000	1000
2	SPE Cartridge Equilibration	5% Ammonium Hydroxide	5000	1000
3	Load Samples onto MAX Cartridge	5% Ammonium Hydroxide	2000	1000
4	Wash MAX Cartridge	5% Ammonium Hydroxide	5000	1000
5	Sample Elution onto Analytical Column	Water/0.1% FA	200	200

Table 3. Solid-phase hysphere GP extraction conditions

#	Step	Solvent	Flow Rate (µL/min)	Volume (µL)
1	SPE Cartridge Activation	ACN/0.1% TFA	5000	1000
2	SPE Cartridge Equilibration	Water/0.1% TFA	5000	1000
3	Load Samples onto GP Cartridge	Water/0.1% TFA	2000	1000
4	Wash GP Cartridge	Water/0.1% TFA	5000	1000
5	Sample Elution onto Analytical Column	Water/0.1% FA	200	200

Results

- Preliminary results demonstrated that MAX extraction provided "cleaner" sample preparation
- When the same volume of plasma sample was injected onto the system, the plasma background (indicated by levels of observed phospholipids) for MAX was significantly lower than that observed using GP cartridges; however, the absolute signal-to-noise for the analyte was higher for GP versus MAX extraction
- In addition, using 0.1% of acid-cleavable PPS surfactant provided effective quenching of enzymatic activity as well as improved extraction recovery without compromising mass spectral quality
- The surfactant enhanced recovery of large hydrophobic peptides by reducing peptide interaction with various plasma proteins and by increasing solubility
- Use of PPS surfactant produced a higher % CV for sample recovery than that observed when no surfactant was used*
- More investigation is necessary to understand the nature of the increased % CV

*This observation is exclusive to GP cartridges and is not observed with MAX cartridges

Table 4. Signal intensity calculation of GP and MAX extractions

	Analyte _Peak Area Count	Plasma Background*_Peak Area Count	Ratio of Analyte/Plasma Background*
GP	3.41E+06	1.55E+05	22
MAX	3.88E+05	2.49E+03	155.8
Ratio of GP/MAX	8.8	62.2	

*As indicated by phospholipids peak.

Figure 1. Mass spectra obtained by using Oasis MAX and Hysphere GP extractions

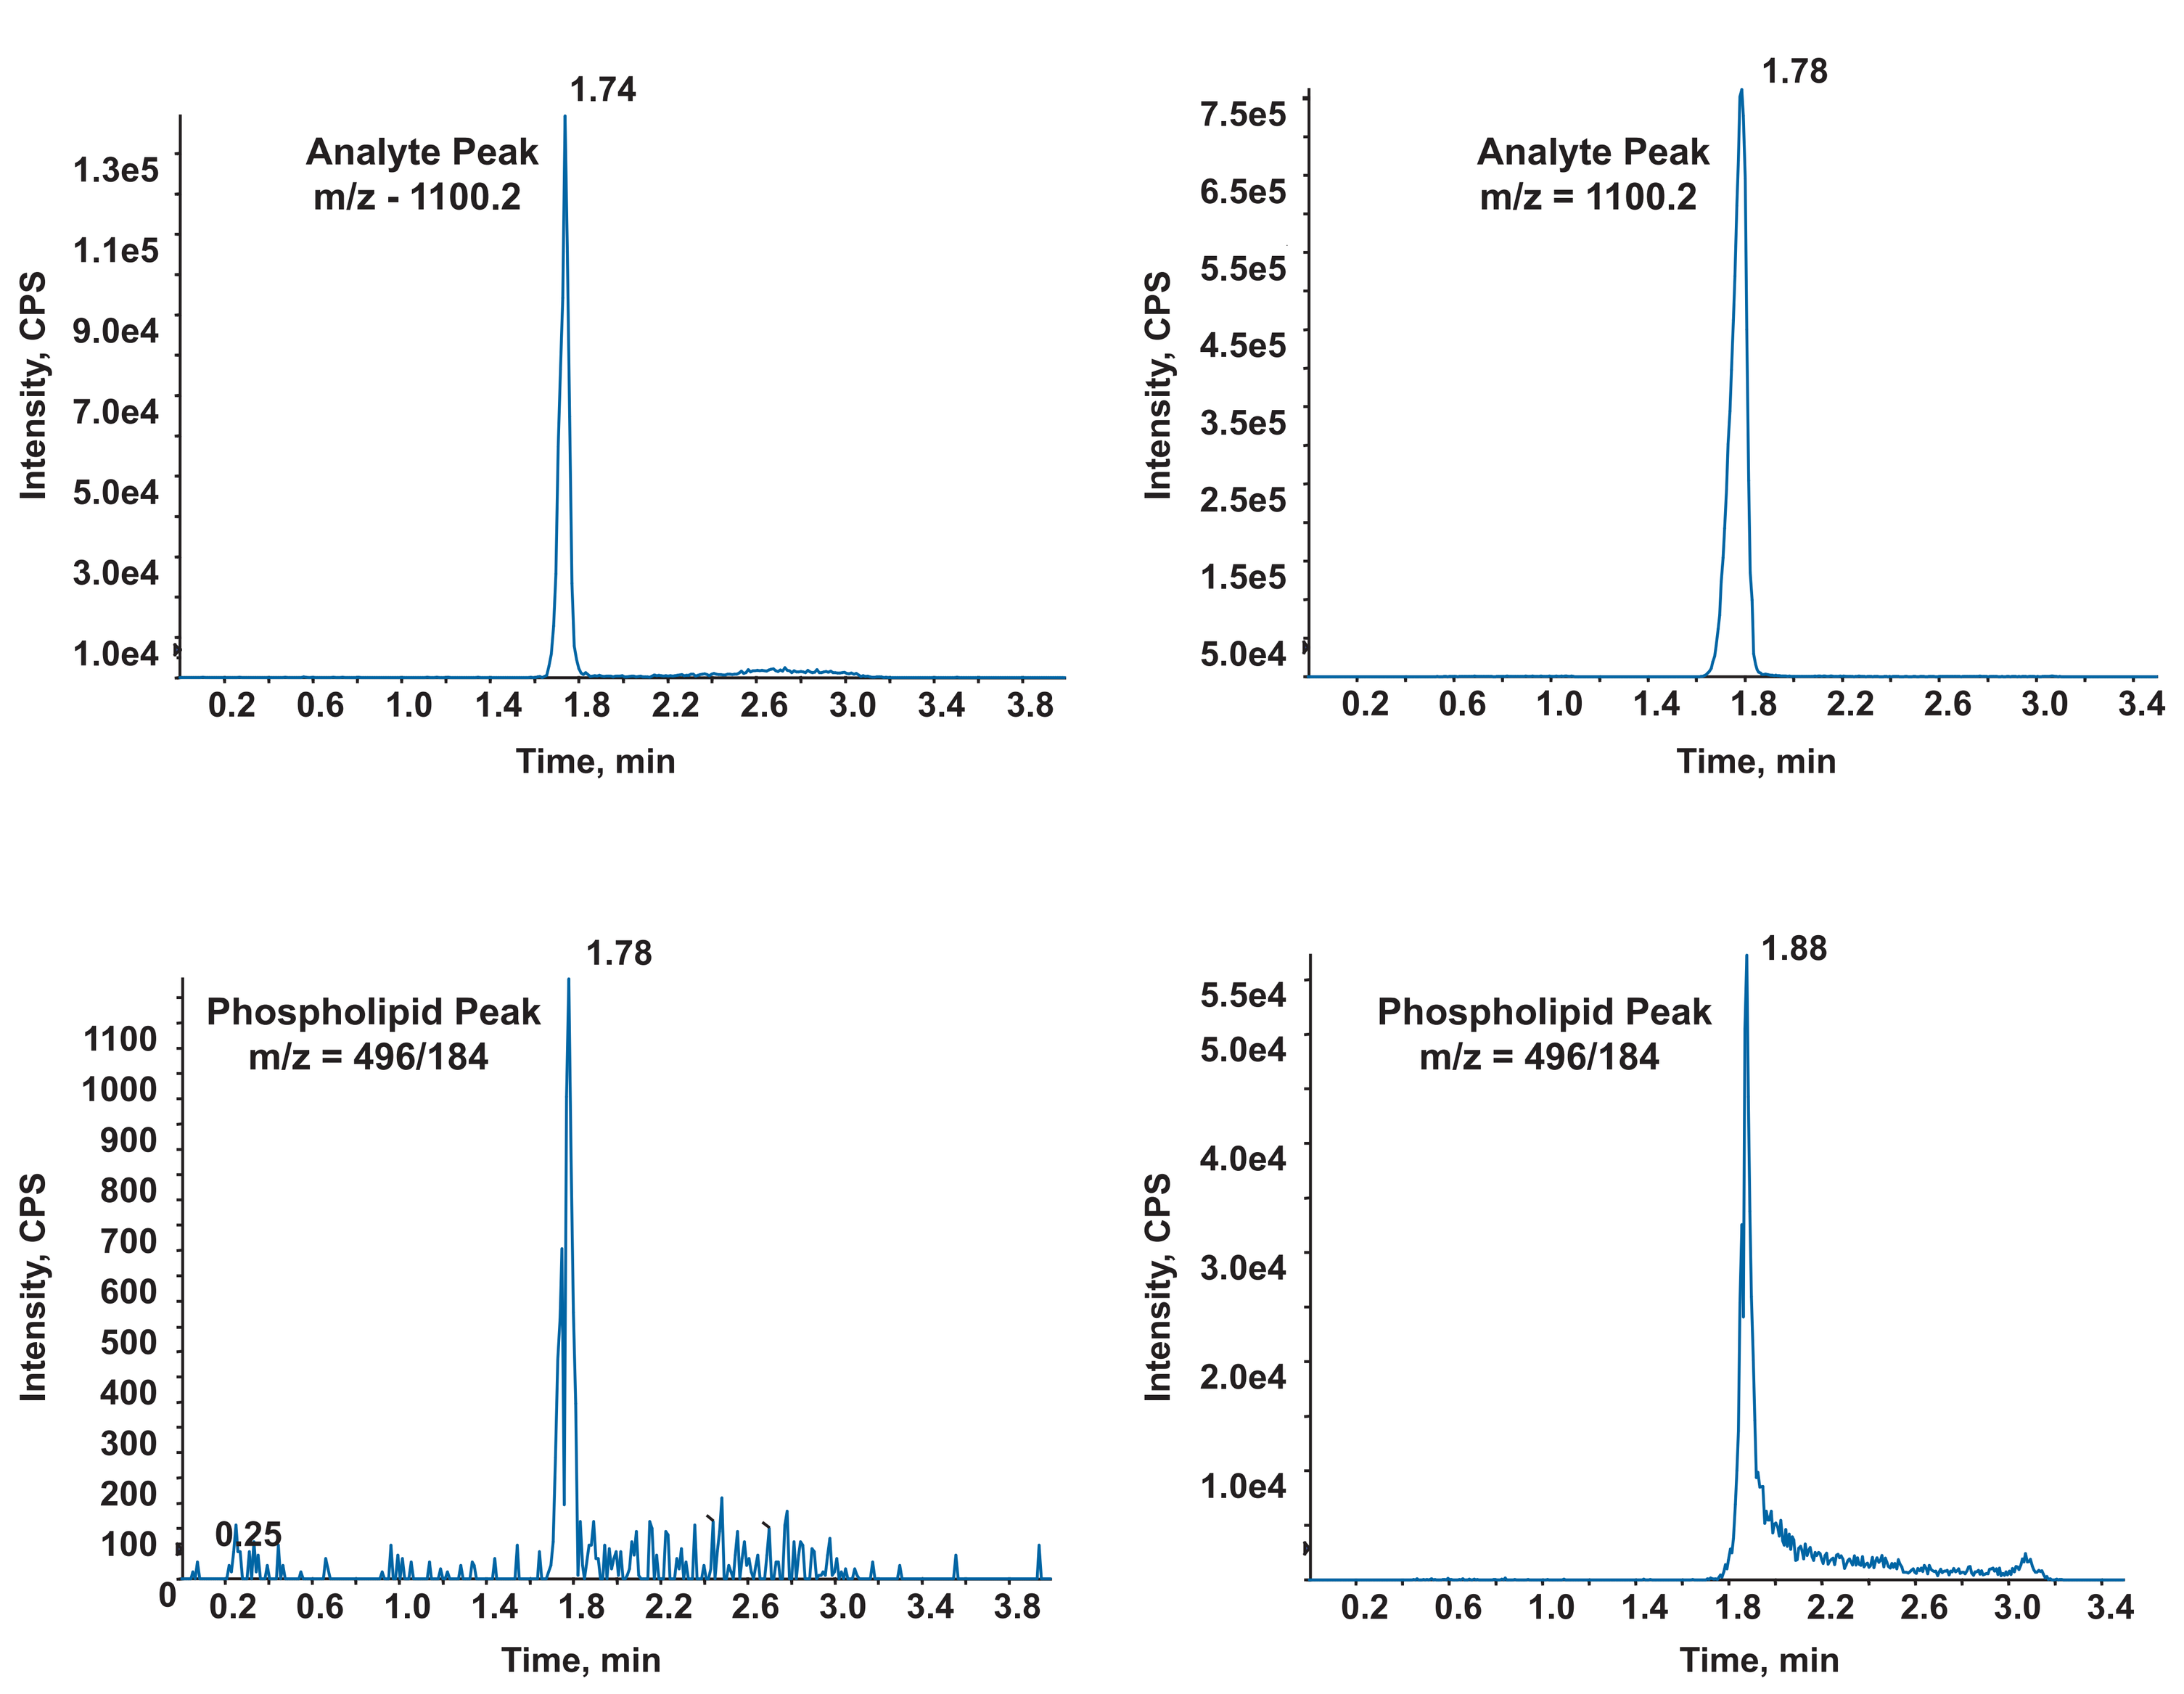
MAX Extraction
GP Extraction
Analyte Peak
m/z - 1100.2
Analyte Peak
m/z = 1100.2
1.74
1.78
Phospholipid Peak
m/z = 496/184
1.78
1.88
0.25
Intensity, CPS
Time, min

Figure 2. Effects of PPS concentrations

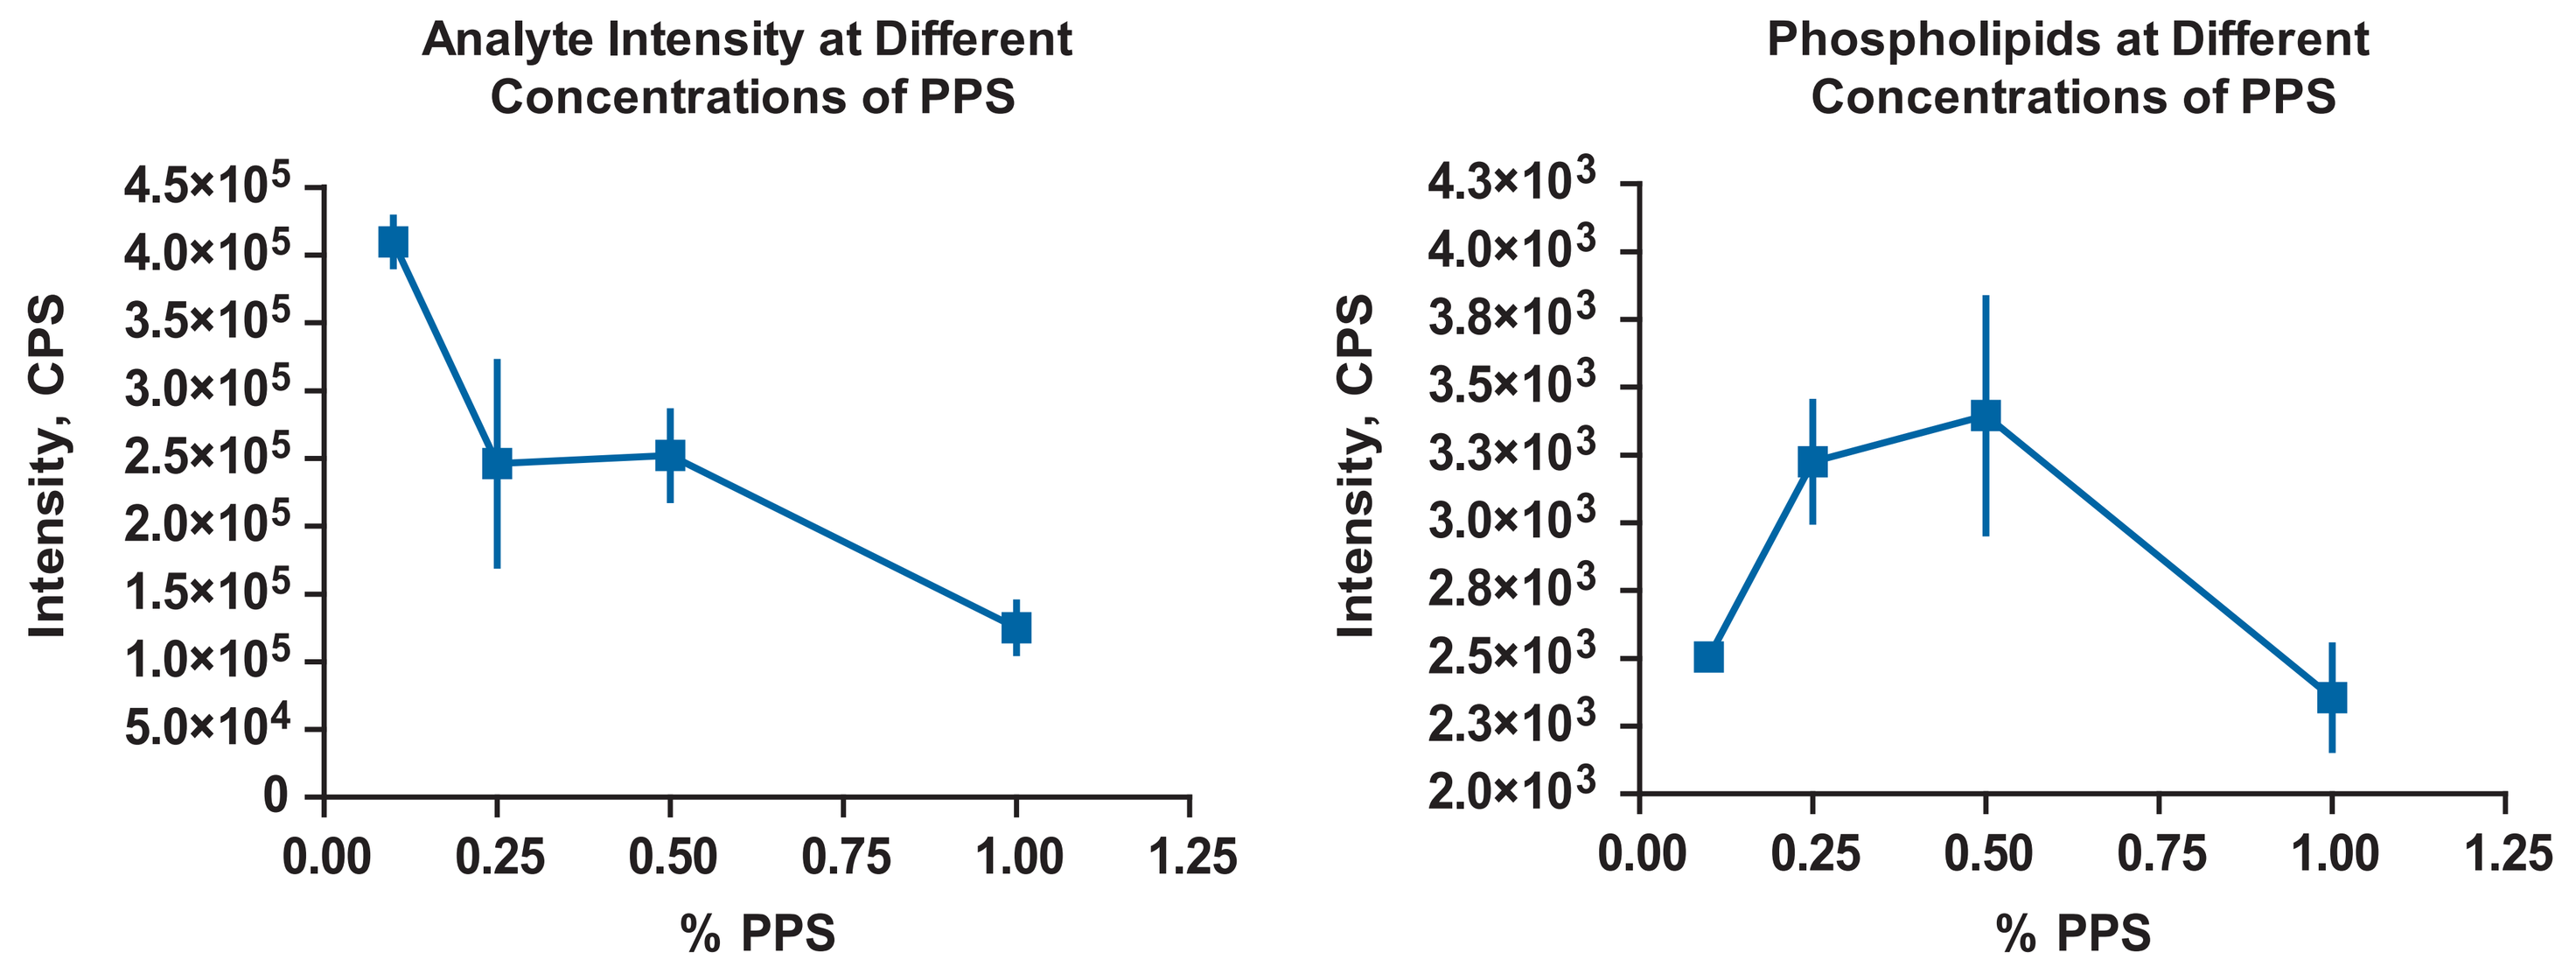
Analyte Intensity at Different Concentrations of PPS
Phospholipids at Different Concentrations of PPS
Intensity, CPS
% PPS

PPS concentration tested: 0.1; 0.25; 0.5, and 1.0%.
Analyte: A peptide in human plasma.
Results: Highest extraction recovery (analyte/plasma) was found with 0.1% PPS application.

Comparisons of PPS, NH_4OH, and FA as Diluents in MAX Sample Preparation

- Samples were prepared by adding a peptide to pooled human plasma
- Prior to MAX cartridge extraction, plasma samples were diluted 1:4 with PPS (final concentration 1%, NH_4OH (final concentration 5%), or FA (final concentration 1%)
- 50 µL of fresh diluted samples (0.3 h) was injected onto a Symbiosis Pharma™ system and subsequently eluted from the MAX cartridge onto a reverse-phase analytical column coupled to an API 3000 mass spectrometer system; API 3000 mass spectrometer provided SIM detection
- Same amount of each sample was analyzed using same instrumental system and method, after incubation at room temperature for 5 (5 h) or 15 (15 h) hours
- Conclusions: after 15 h, 83% of analyte signal (compare to fresh sample) was detected in the samples with 0.1% of PPS; Only 65% or 49% of intensity remained in samples with 5% of NH_4OH or 1% FA, respectively

Figure 3. Comparison of PPS, NH_4OH, and FA as diluents using MAX cartridges

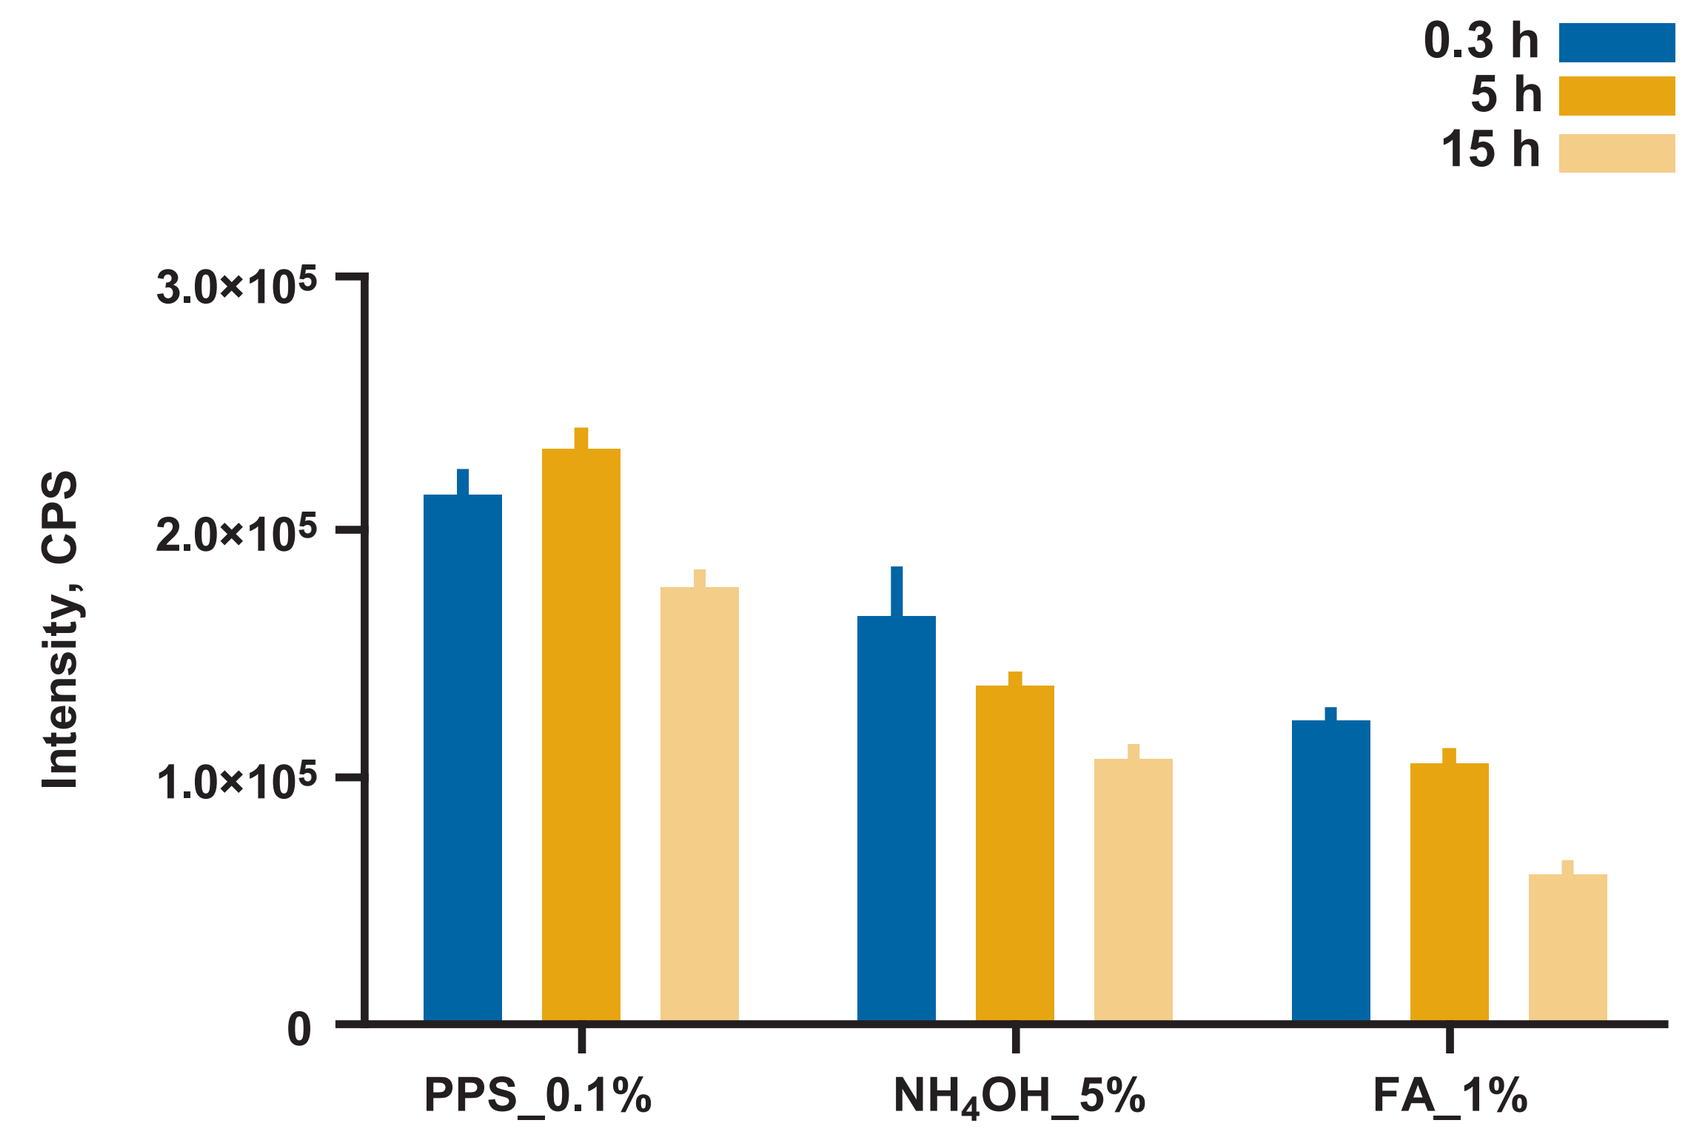
0.3 h
5 h
15 h
Intensity, CPS
PPS_0.1%
NH4OH_5%
FA_1%

Comparisons of PPS, NH_4OH, FA, and H_3PO_4 as Diluents in GP Sample Preparation

- Samples were prepared by adding a peptide to pooled human plasma
- Prior to GP cartridge extraction, plasma samples were diluted 1:4 with PPS (final concentration 0.1%), NH_4OH (final concentration 5%), FA (final concentration 1%), or H_3PO_4 (final concentration 5%)
- 50 µL of fresh diluted samples (0.3 h) was injected onto a Symbiosis Pharma system and subsequently eluted from the GP cartridge onto a reverse-phase analytical column coupled to an API 4000 mass spectrometer system, which provided SIM detection
- The same amount of each sample was analyzed using same instrumental system and method, after incubation at room temperature for 5 (5 h) or 15 (15 h) hours
- Conclusions: after 15 h, almost 100% of analyte signal (compare to fresh sample) was detected in the samples with all 4 different diluents tested; however, the STDEV was higher for plasma samples diluted with PPS

Figure 4. Comparison of PPS, NH_4OH, FA, and H_3PO_4 as diluents using GP cartridges

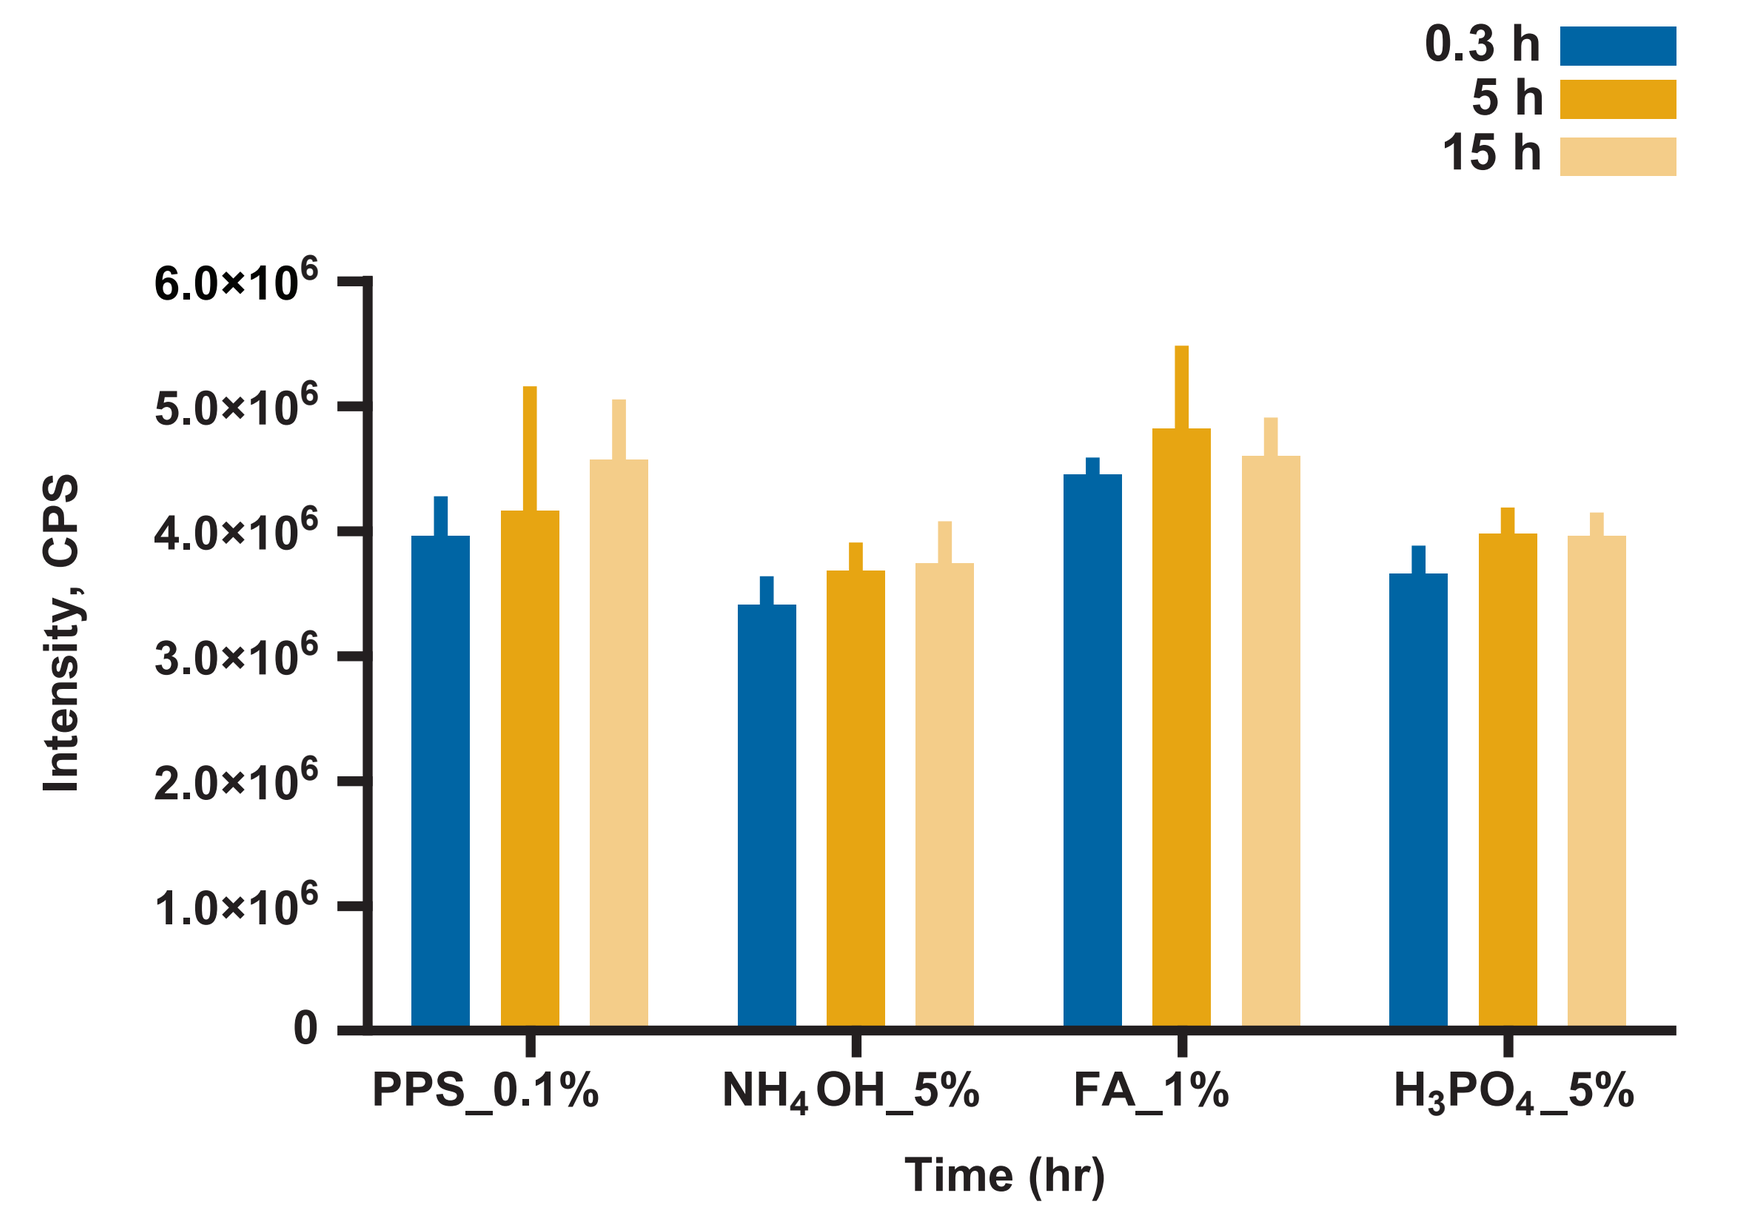
0.3 h
5 h
15 h
Intensity, CPS
PPS_0.1%
NH4OH_5%
FA_1%
H3PO4_5%
Time (hr)

Conclusion

- Anion exchange MAX cartridge provided "cleaner" sample preparation
- When the same amount of plasma samples were injected onto the system, the plasma background for MAX was significantly lower than that of GP cartridges
- Preliminary results showed that the signal intensity of analyte/plasma background was 156 and 22 for MAX and GP, respectively; however, the absolute signal-to-noise for the analyte was higher for GP extraction (3.41e6) versus MAX (3.88e5)
- PPS at concentration of 0.1% provided effective quenching of enzymatic activity as well as improved extraction recovery without compromising mass spectral quality
- PPS enhanced recovery of large hydrophobic peptides by reducing peptide interaction with various plasma proteins and by increasing solubility; however, our data showed that the % CV was higher for plasma samples diluted with PPS when using GP cartridge extraction
- More investigation to understand the nature of the increased % CV is necessary